Application of Capillary Isoelectrofocusing for protein analysis

Marina Kirkitadze, Jian Hu, Matthew Todd, and George Cates

Research Biochemistry, Sanofi Pasteur, Toronto, Canada

Introduction

Capillary isoelectric focusing (cIEF) is an analytical method that allows detection of heterogeneity in the sample related to glycosylation and impurities, and offers many advantages over gel-based isoelectric focusing (IEF) such as automation, quantitation and faster analysis speed. The analysis of antigen's stability and consistency is a critical component of the manufacturing process that requires a variety of characterization methods capable to determine similarities and differences related to process. To expand characterization package cIEF method was developed to analyze the native, homologous, and chemically modified protein antigens.

Methods

Principle and Purpose of the Method

IEF is an electrophoretic method that separates proteins according to their isoelectric points (pI). In a pH gradient, under the influence of an electric field, a protein will move to the position in the pH gradient where its net charge is zero. A protein with a positive net charge will move to the cathode, becoming progressively less positively charged as it moves through the pH gradient until it reaches its pI. A protein with a negative net charge will on the other hand move towards the anode until it reaches its pI. This focusing effect concentrates proteins at their pI and allows proteins to be separated on the basis of very small charge differences. Capillary isoelectrofocusing provides a higher degree of resolution compared to gels due to a higher focusing voltage and shorter time of focusing

Procedure

Isoelectrofocusing was done using a capillary imaged analyzer iCE280 (Convergent Biosciences, Toronto, Canada). iCE280 incorporates whole-column detection to monitor the IEF peak pattern of a protein within a capillary column. The IEF capillary column was conditioned using 0.5 % methylcellulose (MC) for 2 hours as described in iCE280 Analyzer Operator's Guide. Test samples were mixed with 0.5 % MC and injected into the capillary column using autosampler. For each run, the injected sample volume was 40 µL. The sample focusing times were 3 to 5 min. The anolyte solution was 0.08 M H3PO4 with 0.1 % MC. The catholyte solution was 0.1 M NaOH with 0.1 % MC. To measure pI a mixture of synthetic pI markers (pI 4.4 and 7.2) were injected with the sample (Bio Rad, Hercules, CA, USA) as internal reference standards. iCE280 CFR software 3.0 was used to acquire and analyze the spectra.

Figure 1 : Schematic diagram

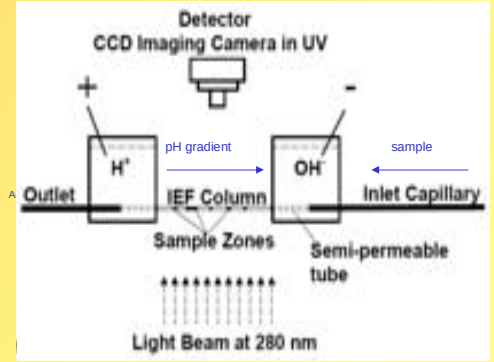

Detector
CCD Imaging Camera in UV
+
-
pH gradient
sample
H+
OH-
Outlet
IEF Column
Inlet Capillary
Sample Zones
Semi-permeable tube
Light Beam at 280 nm

Courtesy of: Jiaqi Wu, Convergent Bioscience

Protein N lot-to-lot consistency

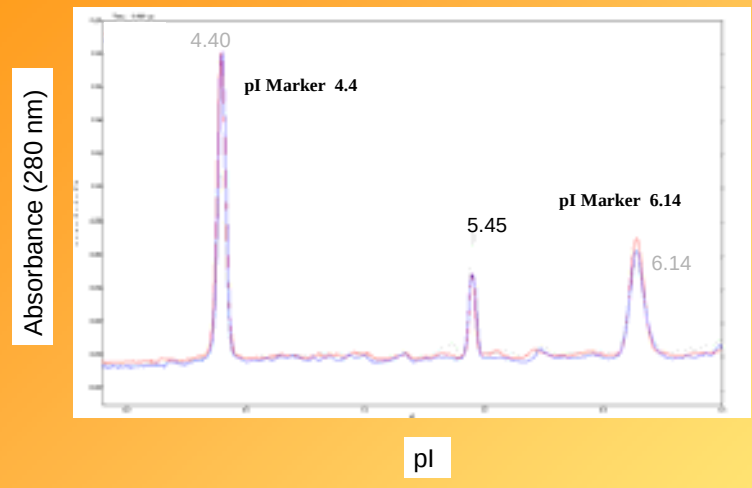

Absorbance (280 nm)
4.40
pI Marker 4.4
5.45
pI Marker 6.14
6.14
pI

Three lots of Protein N were analyzed by cIEF to determine process consistency. DSC was used in parallel to monitor thermal stability which was similar for all three lots

cIEF spectra of chemically treated Protein Q

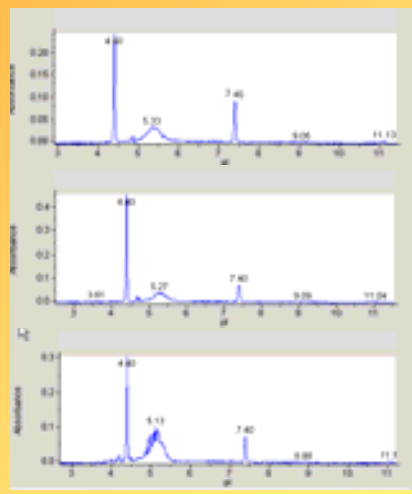

Process 1

Process 2

Process 3

Protein Q produced using three different processes was compared by CD, DSC, and cIEF. All three samples had similar helical profile. No difference was detected by DSC for Protein Q obtained by processes 1 and 2, however showed lower transition temperature for that of process 3. cIEF detected subtle differences between the samples

Protein X non-tagged and His-tagged

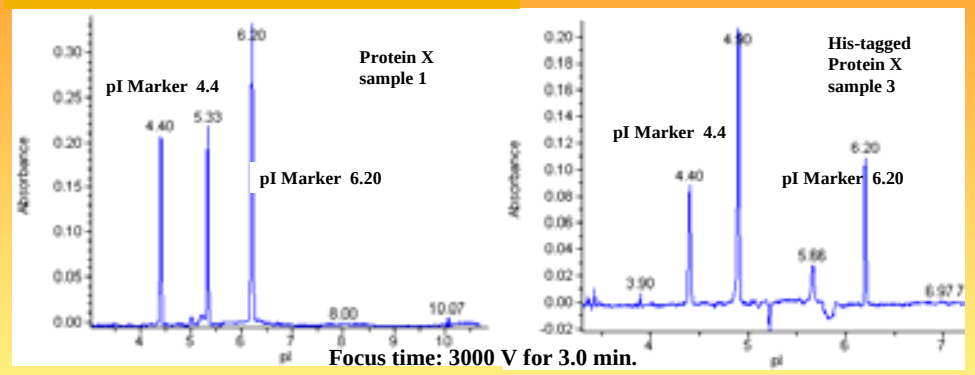

pI Marker 4.4
Protein X sample 1
pI Marker 6.20
4.40
5.33
6.20
8.00
10.07
Absorbance
His-tagged Protein X sample 3
pI Marker 4.4
pI Marker 6.20
3.90
5.86
Focus time: 3000 V for 3.0 min.

His-tagged Protein X appears to be more acidic than a non-tagged one. This observation was consistent with lower thermal stability of His-tagged protein shown by DSC. CD and AUC showed no differences in secondary structure and stoichiometry of the proteins.

cIEF distinguishes between homologous proteins

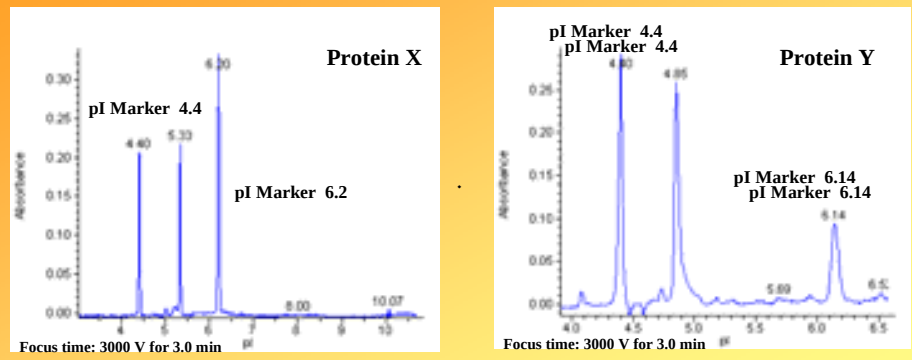

Protein X
pI Marker 4.4
pI Marker 6.2
Focus time: 3000 V for 3.0 min
pI Marker 4.4
pI Marker 4.4
Protein Y
pI Marker 6.14
pI Marker 6.14
Focus time: 3000 V for 3.0 min

Homologous proteins X and Y have helical secondary structure, monomeric form, and similar thermal stability, as shown by DSC, CD, and AUC. However, they can be distinguished by their pI values

Summary

cIEF allows to:

Determine pI of protein in solution

Distinguish between homologous proteins

Monitor process consistency and stability

Obtain signature profile for complex mixtures

References

Citations

1. Wu, J. Li, S-C., Watson, A. (1998). Optimizing separation conditions for proteins and peptides using imaged capillary isoelectric focusing method. Journal of Chromatography A, 817: 163-171.
2. Wu, J., Huang, T., Determination of Isoelectric Point (pI) by Whole-Column Detection cIEF. Convergent Bioscience. Accessed: 31 August 2006. http://www.ciefforum.com/download_file.php?id=pdf/CEpharm05poster.pdf
3. Righetti, P.G., (2004). Determination of the isoelectric point of proteins by capillary isoelectric focusing. Journal of Chromatography A, 1037: 491-499.
4. Wu, J., Improving IEF's Analytical Success Rate for Biopharmaceutical Samples. Convergent Bioscience. Accessed: 31 August 2006. http://www.ciefforum.com/download_file.php?id=pdf/TechSeminar3.pdf
5. Wu, J., A Practical Guide to Capillary Isoelectric Focusing (cIEF) Method Development. Convergent Bioscience. Accessed: 31 August 2006. http://www.ciefforum.com/download_file.php?id=pdf/Technical%20Seminar%202.pdf
6. Convergent Bioscience, iCE 280 Analyzer Frequently Asked Questions. Convergent Bioscience. Accessed: 31 August 2006. http://www.convergentbiosci.com/support/faq.pdf

Acknowledgements

We would like to thank our colleagues Tao Yuan, Tim Lee, Sandrine Cigarini, Bruce Carpick, Martina Ochs for providing the samples for this study. We also thankful to Jiaqi Wu from Convergent Bioscience for providing support.

sanofi pasteur

The vaccines business of sanofi-aventis Group