Imaging Capillary Isoelectric Focusing of a Humanized Monoclonal Antibody

Methal N. Albarghouthi, John N. Hope, Ziping Wei, and Mark A. Schenerman
Department of Analytical Biochemistry, MedImmune, Gaithersburg, MD 20878 USA

ABSTRACT

A capillary isoelectric focusing (cIEF) method for analysis of a humanized monoclonal antibody using imaging capillary electrophoresis instrument, iCE280 Analyzer, has been developed. Optimal resolution of monoclonal antibody peaks was obtained using a combination of 0.5% (v/v) long range ampholine pH 3.5-10 and 1.5% (v/v) narrow range high-resolution ampholyte pH 8-10. The cIEF method is shown to be accurate for pI determination and specific as it resolved different monoclonal antibodies in a mixed sample based on their pI and peak profiles. The cIEF method exhibited excellent precision for injection repeatability, method repeatability, and day-to-day intermediate precision. The cIEF method was evaluated by demonstrating the robustness and precision of the analysis when the method was subjected to variations in composition of carrier ampholytes, cIEF cartridges, focusing time, and sample volumes, as well as examining the pass/failure rate for the method over 300 experimental runs. The cIEF peak profile was compared to that obtained by cation exchange chromatography (IEC). The relative proportion of C-terminal lysine variants of the monoclonal antibody determined by cIEF and by IEC was found to be comparable. However, when the C-terminal lysine residues were digested by Carboxypeptidase-B, the percentage of the 0-lysine variant peak relative to the total peak profile area was greater in IEC than in cIEF. This is because cIEF exhibited a better overall resolution of the peaks that are more acidic than the major 0-lysine peak. The study demonstrates the applicability of cIEF as a complement to IEC for the characterization of charge heterogeneity of monoclonal antibodies.

INTRODUCTION

Isoelectric focusing (IEF) has been routinely used in biotechnology to monitor purity, stability, and heterogeneity of therapeutic monoclonal antibodies. Compared to slab gel format, capillary isoelectric focusing (cIEF) provides high-resolution, quantitative analysis, short run time, online sample detection, and fully automated operation. In cIEF, a capillary is filled with a protein sample mixed with carrier ampholytes. An electric field is then applied to the capillary resulting in a pH gradient generated by the carriers ampholytes. Protein molecules are focused in the pH gradient according to their isoelectric points. Next, the focused protein bands are then typically mobilized past a detector, using either chemical or pressure mobilization, which can distort resolution of the bands, and increase analysis time. The iCE280 Analyzer (Convergent Biosciences) utilizes whole-column imaging technology, which eliminates the need for mobilizing focused protein bands past a detector, thus avoiding the problems associated with this process.

The purpose of this study is to develop a cIEF method using iCE280 Analyzer for analysis of a humanized monoclonal antibody (MAb). The method is optimized with respect to ampholyte type and composition to yield the best resolution of peaks. The study evaluates the accuracy, specificity, precision, and robustness of the cIEF method using the iCE280 Analyzer. Additionally, the study examines the suitability of the method for use as a stability-indicating assay for MAb samples. Then, the performance of the cIEF and ion exchange chromatography (IEC) techniques are compared for quantitative evaluation of lysine variants and of peaks that are more acidic than the 0-lysine variant peak.

MATERIALS AND METHODS

cIEF Method: cIEF separations were performed in a 100 mm ID, 5 cm FC cIEF cartridge. Samples were prepared by mixing 15ul of 10 mg/ml protein sample with 200ul of 2% ampholyte mixture composed of 25% broad range Ampholine pH 3.5-10 (Sigma-Aldrich) and 75% narrow range high-resolution ampholyte pH 8-10 (Fluka) in 0.5% methyl cellulose solution. Anode and cathode solutions were 100 mM phosphoric acid, and 80 mM sodium hydroxide, in 0.1% methyl cellulose solution. Focusing was performed at 3000 V for 12 minutes. Detection was at 280 nm.

IEC method: MAb samples were injected onto a silica-based Propac™ WCX-10 ion-exchange column at ambient temperature. Mobile phase A is 20 mM sodium phosphate, pH 7.0, mobile phase B is 20 mM sodium phosphate containing 0.1 M sodium chloride, pH 7.0. A gradient of 30% - 60% mobile phase B over 40 minutes is run at a flow rate of 1.0 mL/min. Eluted protein is detected using UV absorbance at 220 nm.

Phast IEF Gel Method: Protein samples at 10 mg/ml were mixed in a 1:1 ratio with ampholine solution and loaded on a rehydrated Phast IEF gel (Pharmacia Biotech). The gel was initially focused for 1 hour at 500 V/cm then at 1000 V/cm for an additional 45 minutes. Then the gel was stained with Coomassie blue.

Identification of Terminal Lysine Variants: Carboxypeptidase B (CBP) was used to cleave the C-terminal lysine residues in the MAb. Cleavage was carried out by incubating a protein sample with 2 ml of CBP solution at 37 °C for 15-30 min. For identification of lysine variants, a MAb sample digested with CBP is compared to the undigested (control) sample. The samples are analysed by ion exchange chromatography. The percent of variants 0-Lys (no lysines on C-terminus of either H chain), 1-Lys (one lysine on C-terminus of either H chain), and 2-Lys (lysine on C-terminus of both H chains) peaks are calculated based on their peak areas.

RESULTS

Effect of Carrier Ampholyte Type

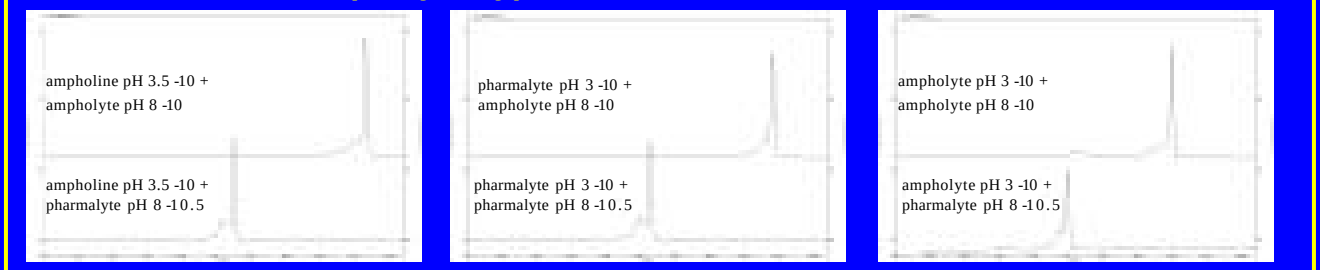
ampholine pH 3.5-10 +
ampholyte pH 8-10
ampholine pH 3.5-10 +
pharmalyte pH 8-10.5
pharmalyte pH 3-10 +
ampholyte pH 8-10
pharmalyte pH 3-10 +
pharmalyte pH 8-10.5
ampholyte pH 3-10 +
ampholyte pH 8-10
ampholyte pH 3-10 +
pharmalyte pH 8-10.5

Figure 1: Comparison of cIEF profiles of MAb using different combinations of long and narrow range carrier ampholytes.

The best resolution of MAb peaks was obtained using ampholyte pH 8-10 in combination with either ampholine pH 3.5-10 or pharmalyte pH 3-10. Ampholyte pH 3-10 gave a comparable resolution but produced disturbance in the baseline. The use of pharmalyte pH 8-10.5 could not produce the same resolution of peaks.

Accuracy

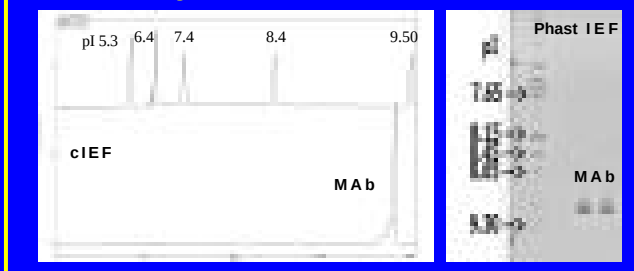
pI 5.3
6.4
7.4
8.4
9.50
cIEF
MAb
Phast IEF
MAb

Figure 2: Determination of pI of main peak of MAb by cIEF (pI = 9.4) and Phast gel IEF (pI = 9.1).

Specificity

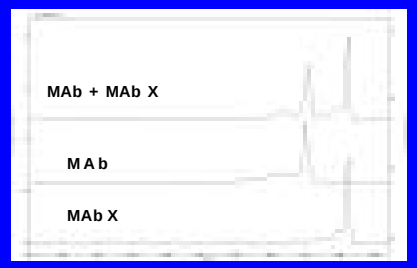
MAb + MAb X
MAb
MAb X

Figure 3: cIEF resolves two MAbs and identifies them according to pI and peak profile.

Instrument Repeatability

Table 1: Instrument repeatability of cIEF of MAb. The %CV of main peak area of six replicates is 1.3%.

Run #	Main peak position (pixel)	Main peak % area
1	1888	69.4
2	1852	68.3
3	1877	68.1
4	1861	68.6
5	1892	68.9
6	1888	70.6
Average	**1876**	**69**
% CV	**0.9**	**1.3**

Method Repeatability

Table 2: Method repeatability of cIEF analysis of MAb. The %CV of main peak area of three samples is 0.5%.

Sample #	Main peak position (pixel)	Main peak % area
1	1932	69.3
2	1936	70
3	1948	69.9
Average	**1938**	**69.7**
% CV	**0.4**	**0.5**

Intermediate Precision

Table 3: Intermediate precision of cIEF analysis of MAb. The %CV of main peak area of samples analyzed on three days is 2.1%.

Day #	Main peak position (pixel)	Main peak % area
1	1932	69.3
2	1882	69.2
3	1932	71.8
Average	**1915**	**70.1**
% CV	**1.5**	**2.1**

Robustness Study

Effect of Ampholyte Ratio

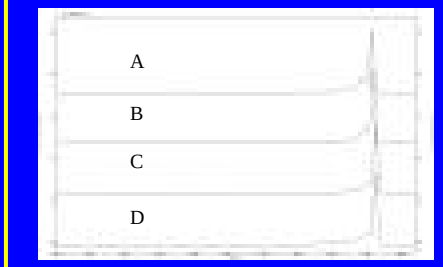
A
B
C
D

Figure 4: cIEF profiles of MAb using ampholine 3.5-10 and ampholyte 8-10 ratios of A. 1.5:0.5, B. 1:1, C. 0.5:1.5, D. 0.2:1.8. Similar peak resolution is obtained using conditions B, C and D. Condition produced poor peak resolution.

Effect of Focusing Time

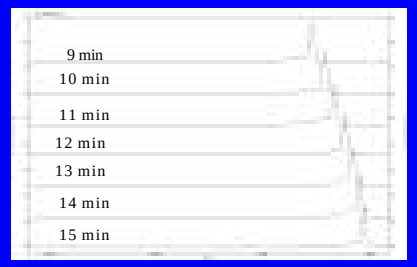
9 min
10 min
11 min
12 min
13 min
14 min
15 min

Figure 5: Effect of focusing time on cIEF of MAb. All MAb peaks are resolved after 11 minutes of focusing.

Effect of sample volume

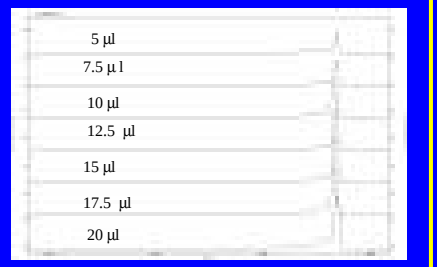
5 µl
7.5 µl
10 µl
12.5 µl
15 µl
17.5 µl
20 µl

Figure 6: Effect of MAb sample volume on cIEF analysis. All MAb peaks are resolved and detected using sample volume ≥ 10 ul.

Cartridge-to-Cartridge Reproducibility

Table 4: Cartridge-to-cartridge reproducibility of MAb cIEF. The %CV of main peak area is 1.0%.

Cartridge #	Main peak position	Main peak % area
1	1932	69.3
2	1894	69.4
3	1902	70.5
Average	**1909**	**69.7**
% CV	**1**	**1**

Table 5: Performance of 300 cIEF runs in three cartridges (100 runs/cartridge). Pass rate and %CVs of each cartridge show robustness of cIEF method.

Cartridge #	Pass rate	Main peak position		Main peak area	
	%	Pixel	%CV	%	%CV
1	91	1913	2	68.8	2.1
2	88	1899	1.6	69.6	2.1
3	96	1923	2.1	69.8	2.2

Detection of Structural Changes in Monoclonal Antibody

The cIEF method was used to detect changes in MAb structure using a heat-stressed MAb sample incubated for 6 months at 38-42 °C. Figure 7 shows a dramatic change in the cIEF peak pattern of the degraded sample compared to the initial condition peak profile. This result demonstrates the applicability of the cIEF method as a stability-indicating assay for MAb.

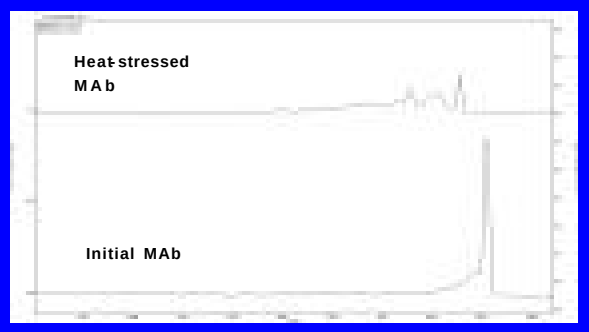
Heat-stressed
MAb
Initial MAb

Figure 7: Change in cIEF profile of heat-stressed MAb.

Determination of Lysine Variants by cIEF and IEC

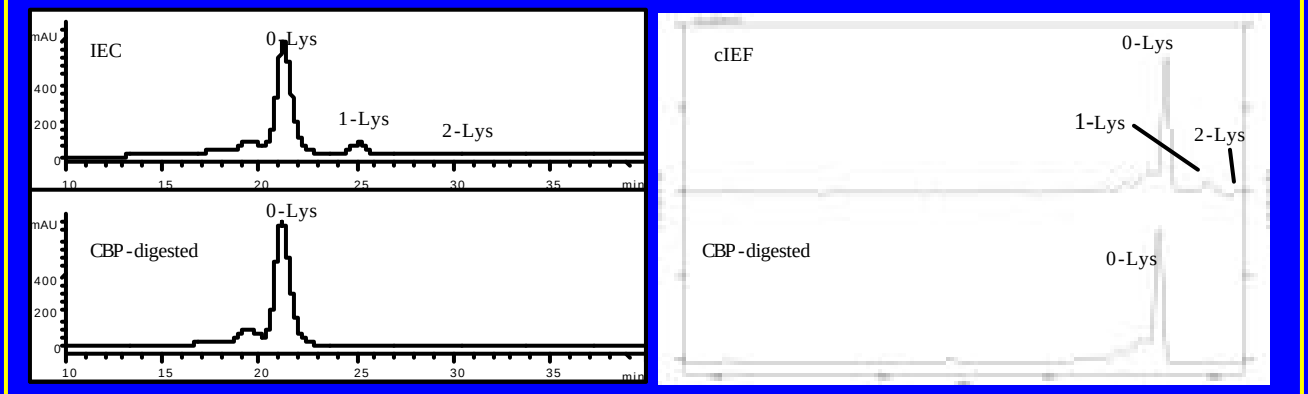
IEC
0-Lys
1-Lys
2-Lys
CBP-digested
0-Lys
cIEF
0-Lys
1-Lys
2-Lys
CBP-digested
0-Lys

Figure 8: Determination of lysine variants of MAb by cIEF and IEC.

Table 4: Relative percentage of lysine variants determined by IEC and cIEF.

Lys variant peak	Peak % area by cIEF	Peak % area by IEC
0-Lys	88	89
1-Lys	11	10
2-Lys	1	1

Table 5: Percentage of 0-Lys variant to total peak area determined by IEC and cIEF in CBP-digested samples from different MAb lots.

MAb Lot	Peak % area by cIEF	Peak % area by IEC
Lot A	60.5	68.2
Lot B	67.8	79.9
Lot C	67.7	78.8

- Figure 8 shows that upon treating MAb with CBP, 2 peaks corresponding to 1-Lys and 2-Lys variants disappeared from the peak profile and co-migrated with the more acidic 0-Lys variant peak. Both cIEF and IEC can resolve single Lys variants of MAb.
- Table 4 shows that the relative percentage of lysine variants determined by IEC and cIEF is consistent
- Table 5 shows that the percentage of the 0-Lys peak in the profile of CBP-digested MAb from three different lots as determined by IEC is higher than that determined by cIEF.
- cIEF provides a better resolution of the peaks that are more acidic than the 0-Lys peak than IEC.

CONCLUSIONS

A cIEF method has been developed for analysis of a monoclonal antibody using a commercially available isoelectric focusing instrument with whole-column imaging capabilities. The cIEF method was shown to be accurate, specific and precise. The robustness of the cIEF method was demonstrated by showing the reproducibility of analysis when subjecting the method to a number of variations in the critical parameters. The pass or failure rate assessment for the cIEF assay over 300 runs also demonstrated the robustness of the method for analyzing MAb. Additionally, the study demonstrated the applicability of the cIEF method as a tool for characterizing charge isoforms of MAbs. Analysis of MAbs by cIEF and IEC produced comparable peak patterns, with cIEF exhibiting a better peak resolution. This work demonstrates the feasibility of employing cIEF as a characterization tool for therapeutic monoclonal antibody analysis.

ACKNOWLEDGEMENTS

We would like to thank Dr. Guillermo Tous for his support of this work, and Jennifer Duehring for providing IEF data.

MedImmune, Inc.